Evaluation of the *In Vitro* Percutaneous Absorption of Ketoprofen Transdermal Formulations

Sara Hover, August S. Bassani, Daniel Banov, and Ha Phan*

PCCA, 9901 South Wilcrest Drive, Houston, TX 77099, USA

PAINWEEK®

Purpose

The purpose of this study is to characterize the percutaneous absorption of 2 ketoprofen formulations (ketoprofen 10% in PLO and ketoprofen 10% in Lipoderm), when applied to the human cadaver trunk skin, *in vitro*, using the Franz skin finite dose model. This model utilizes the finite dose technique and Franz diffusion cells to dose and culture the skin samples. It has been shown to be a valuable tool in predicting *in vivo* percutaneous absorption kinetics of topically applied drugs [1]. Ketoprofen is an analgesic, anti-inflammatory agent that belongs to a class of drugs known as nonsteroidal anti-inflammatory drugs, widely used in the management of chronic musculoskeletal pain [2]. The risk of gastrointestinal complications associated with oral ketoprofen has led to an increase in demand for research and development of ketoprofen formulations for transdermal delivery [3]. Ketoprofen can be incorporated into transdermal bases such as Pluronic Lecithin Organogel (PLO) or Lipoderm, a proprietary phospholipid base, for delivery into and through the skin.

Methods

The percutaneous absorption of ketoprofen was evaluated using human cadaver trunk skin from 3 donors. The skin from each donor was cut into small sections to fit on nominal 1 cm^2 Franz diffusion cells (Fig. 1), chambers specially designed to maintain the skin at a temperature and humidity that match *in vivo* conditions [1]. Within each chamber, the skin section was mounted on a diffusion apparatus so that the dermal layer is immersed in receptor solution while the epidermal surface is exposed to the ambient laboratory condition via the chimney. The receptor solution consisted of 1x-phosphate buffered saline, pH 7.4±0.1, stirred magnetically at approximately 600 revolutions per minute within the receptor compartment. A variable finite dose of each formulation was applied to 3 replicate skin sections per donor, using a positive displacement pipette set to deliver 5 μL formulation/cm^2. At predetermined time points (0, 4, 8, 12, 24, 32, and 48 hr), the receptor solution within each chamber was removed, replaced with fresh receptor solution, and an aliquot was saved for analysis. After the last receptor sample was collected, each skin section was washed to remove unabsorbed formulation from the skin surface. Skin sections were then split into epidermis and dermis, and extracted overnight at room temperature in 50:50 ethanol:water. Ketoprofen content within the receptor solution and the skin were determined via high-performance liquid chromatography analysis of the extractant sample.

Results and Discussion

To characterize the percutaneous absorption of ketoprofen, a total of 3 parameters were determined for each chamber: total absorption, rate of absorption and skin content. Mean values and standard deviation were calculated for each parameter across the 3 donors and expressed as percentages of the applied dose. Total absorption was calculated as the sum of ketoprofen content within the 7 samples collected over 48 hr for each chamber. Rate of absorption, presented as flux (μg/cm^2/hr) of ketoprofen into receptor solution, was determined by dividing the amount of ketoprofen absorbed during a time interval and the length of that interval. Skin content refers to the amount of ketoprofen detected within the dermis and epidermis after 48 hr. The mean total absorption of ketoprofen, when in Lipoderm (14.487%±1.905), was higher than when in PLO (5.513%±1.623). The higher mean total absorption of ketoprofen when in Lipoderm suggests that this base has greater penetration potential than PLO. When examining rate of absorption for both transdermal formulations, mean flux for ketoprofen was higher at each time point, when in Lipoderm, than when in PLO (Fig. 2). Ketoprofen was also detected within the dermis and epidermis. For ketoprofen in PLO, 0.132%±0.037 of the applied dose was found within the dermis and 0.709%±0.131 was in the epidermis. For ketoprofen in Lipoderm, 0.303%±0.115 of the applied dose was detected in the dermis while 1.258% ± 0.066 was in the epidermis. The mean percentage of ketoprofen content detected within the receptor solution (total absorption), dermis, and epidermis were also summed to obtain a mean percent permeation of the applied dose through the stratum corneum. Penetration across the stratum corneum was 6.354% for ketoprofen in PLO and 16.048% for ketoprofen in Lipoderm (Fig. 3). Though results of this study show that PLO and Lipoderm were capable of facilitating the penetration of ketoprofen through the stratum corneum and into underlying layers of the skin, mean percent permeation was higher for the Lipoderm formulation.

Conclusion

Higher mean total absorption, faster rate of absorption and greater penetration across the stratum corneum observed with the ketoprofen in Lipoderm formulation suggest that Lipoderm may be a more appropriate base for the transdermal delivery of ketoprofen in comparison to PLO. Ketoprofen 10% in Lipoderm could potentially offer practitioners and pharmacists a viable alternative option for the management of chronic musculoskeletal pain.

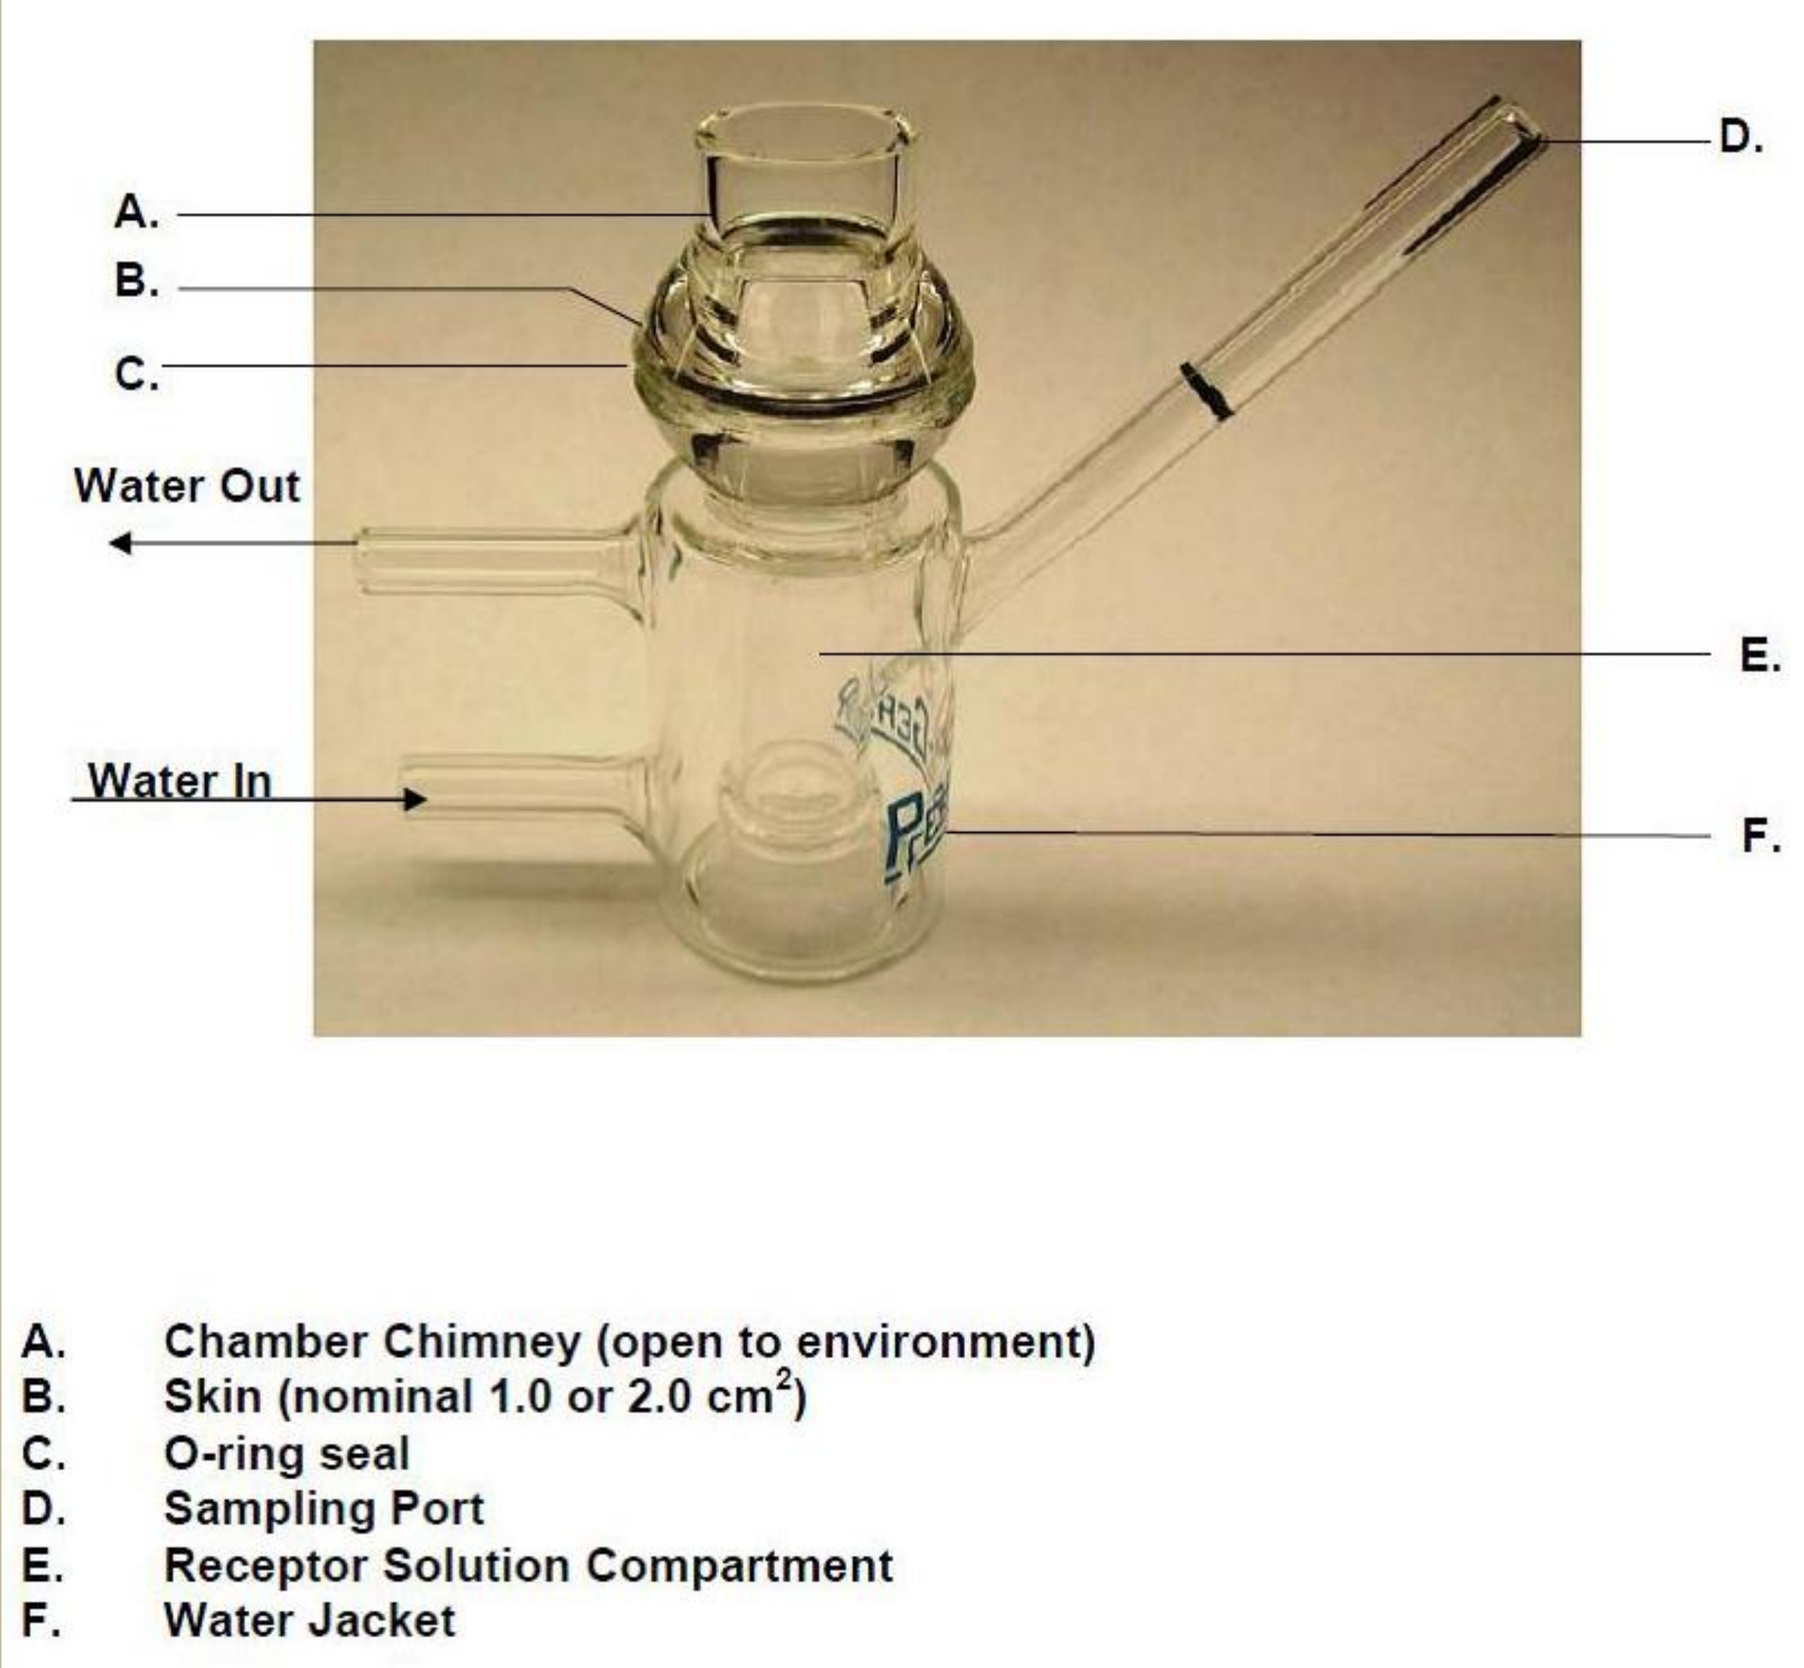
A.
B.
C.
D.
E.
F.
Water Out
Water In
A. Chamber Chimney (open to environment)
B. Skin (nominal 1.0 or 2.0 cm2)
C. O-ring seal
D. Sampling Port
E. Receptor Solution Compartment
F. Water Jacket

Figure 1.
Franz diffusion cell

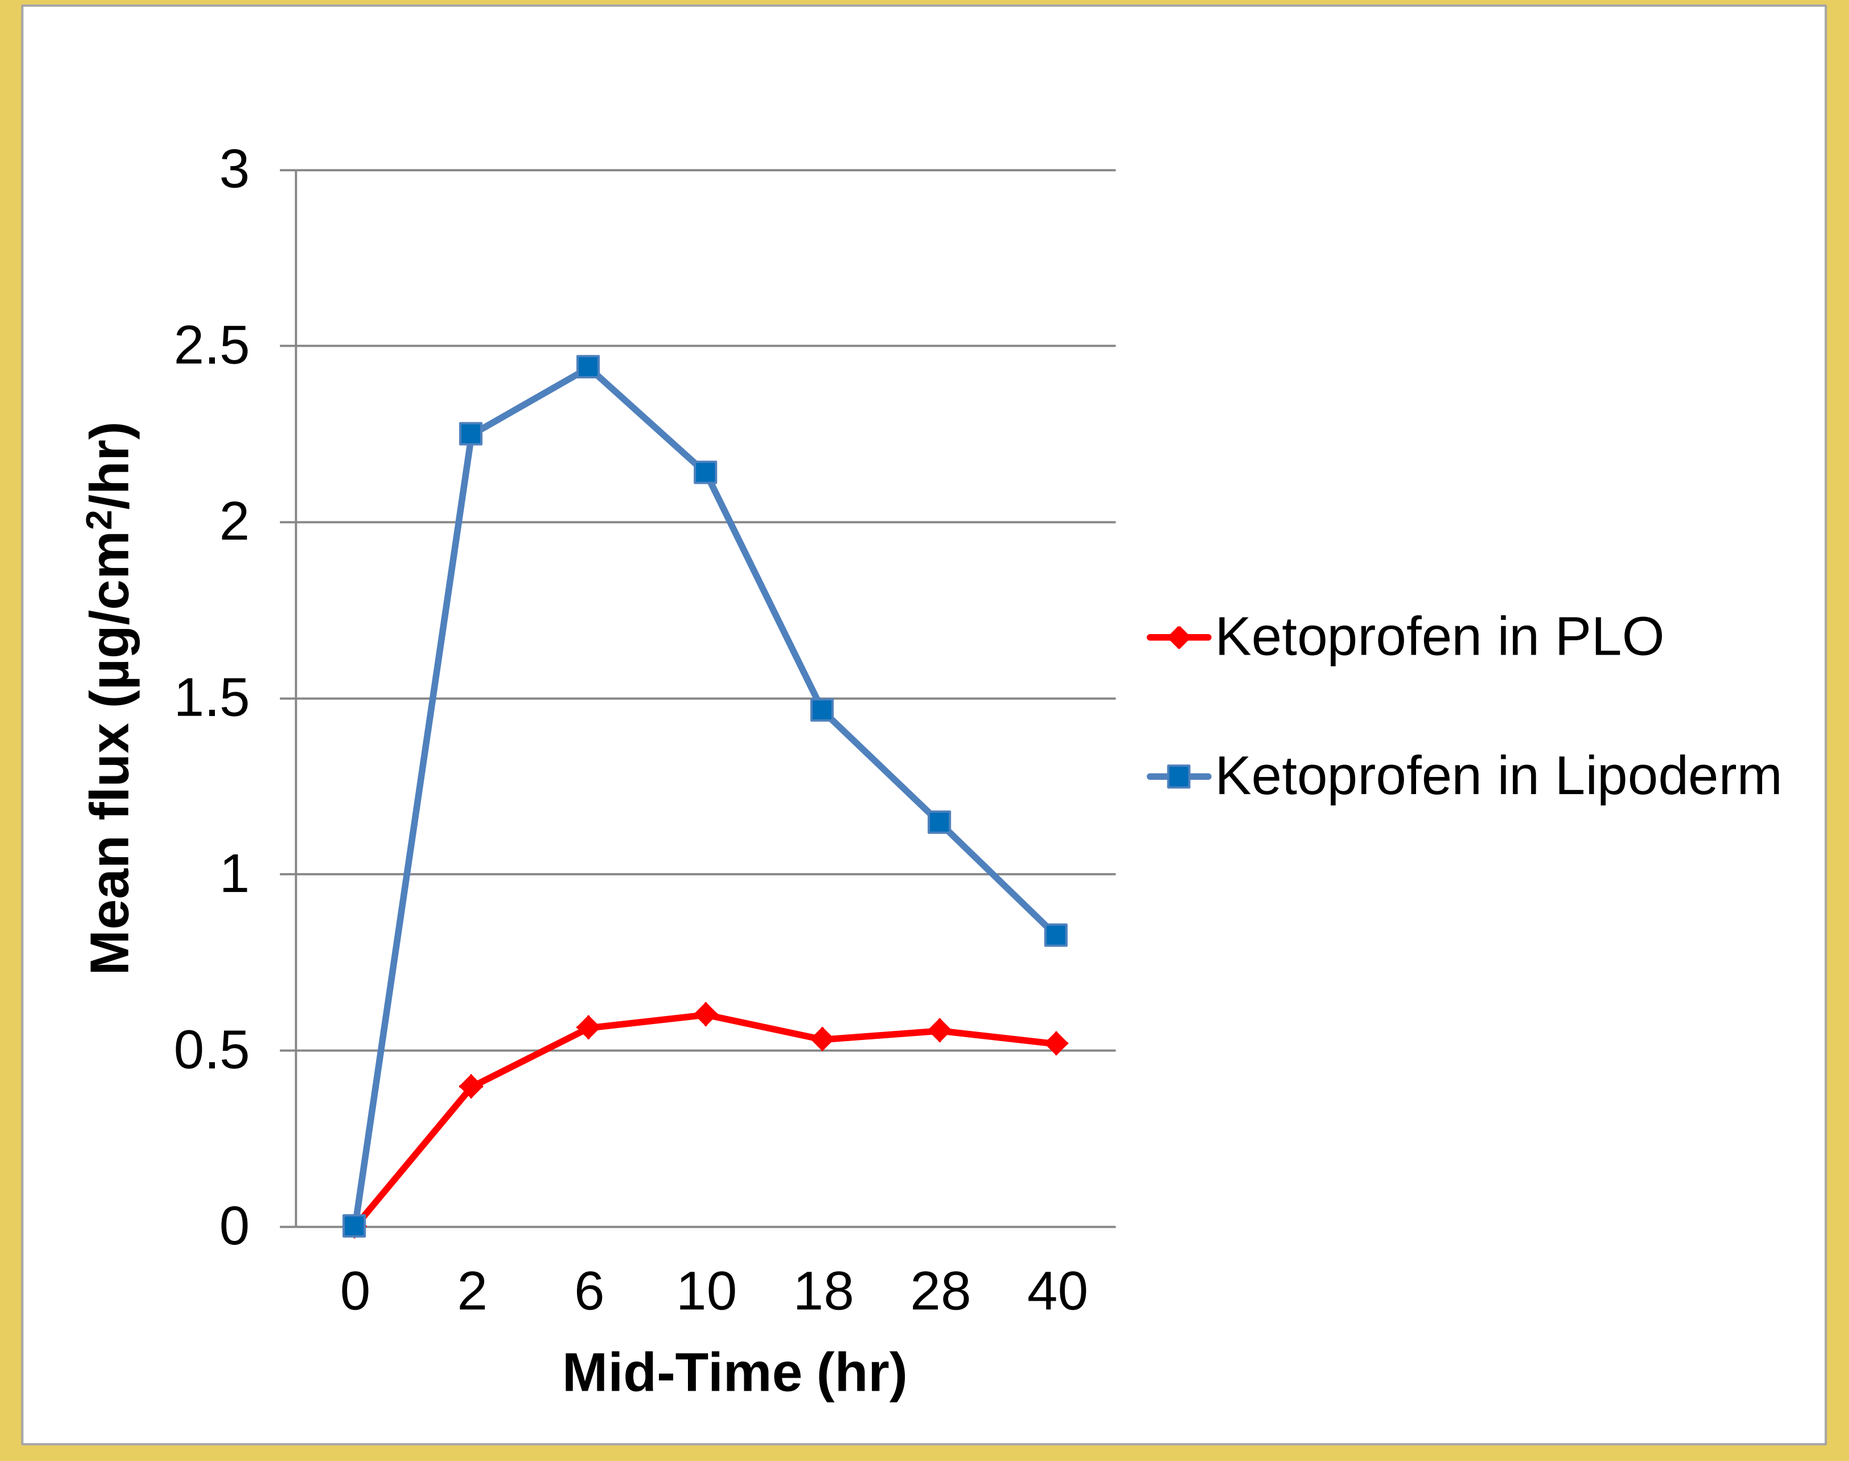
3
2.5
2
1.5
1
0.5
0
0
2
6
10
18
28
40
Mid-Time (hr)
Mean flux (µg/cm²/hr)
Ketoprofen in PLO
Ketoprofen in Lipoderm

Figure 2.
Ketoprofen flux versus time

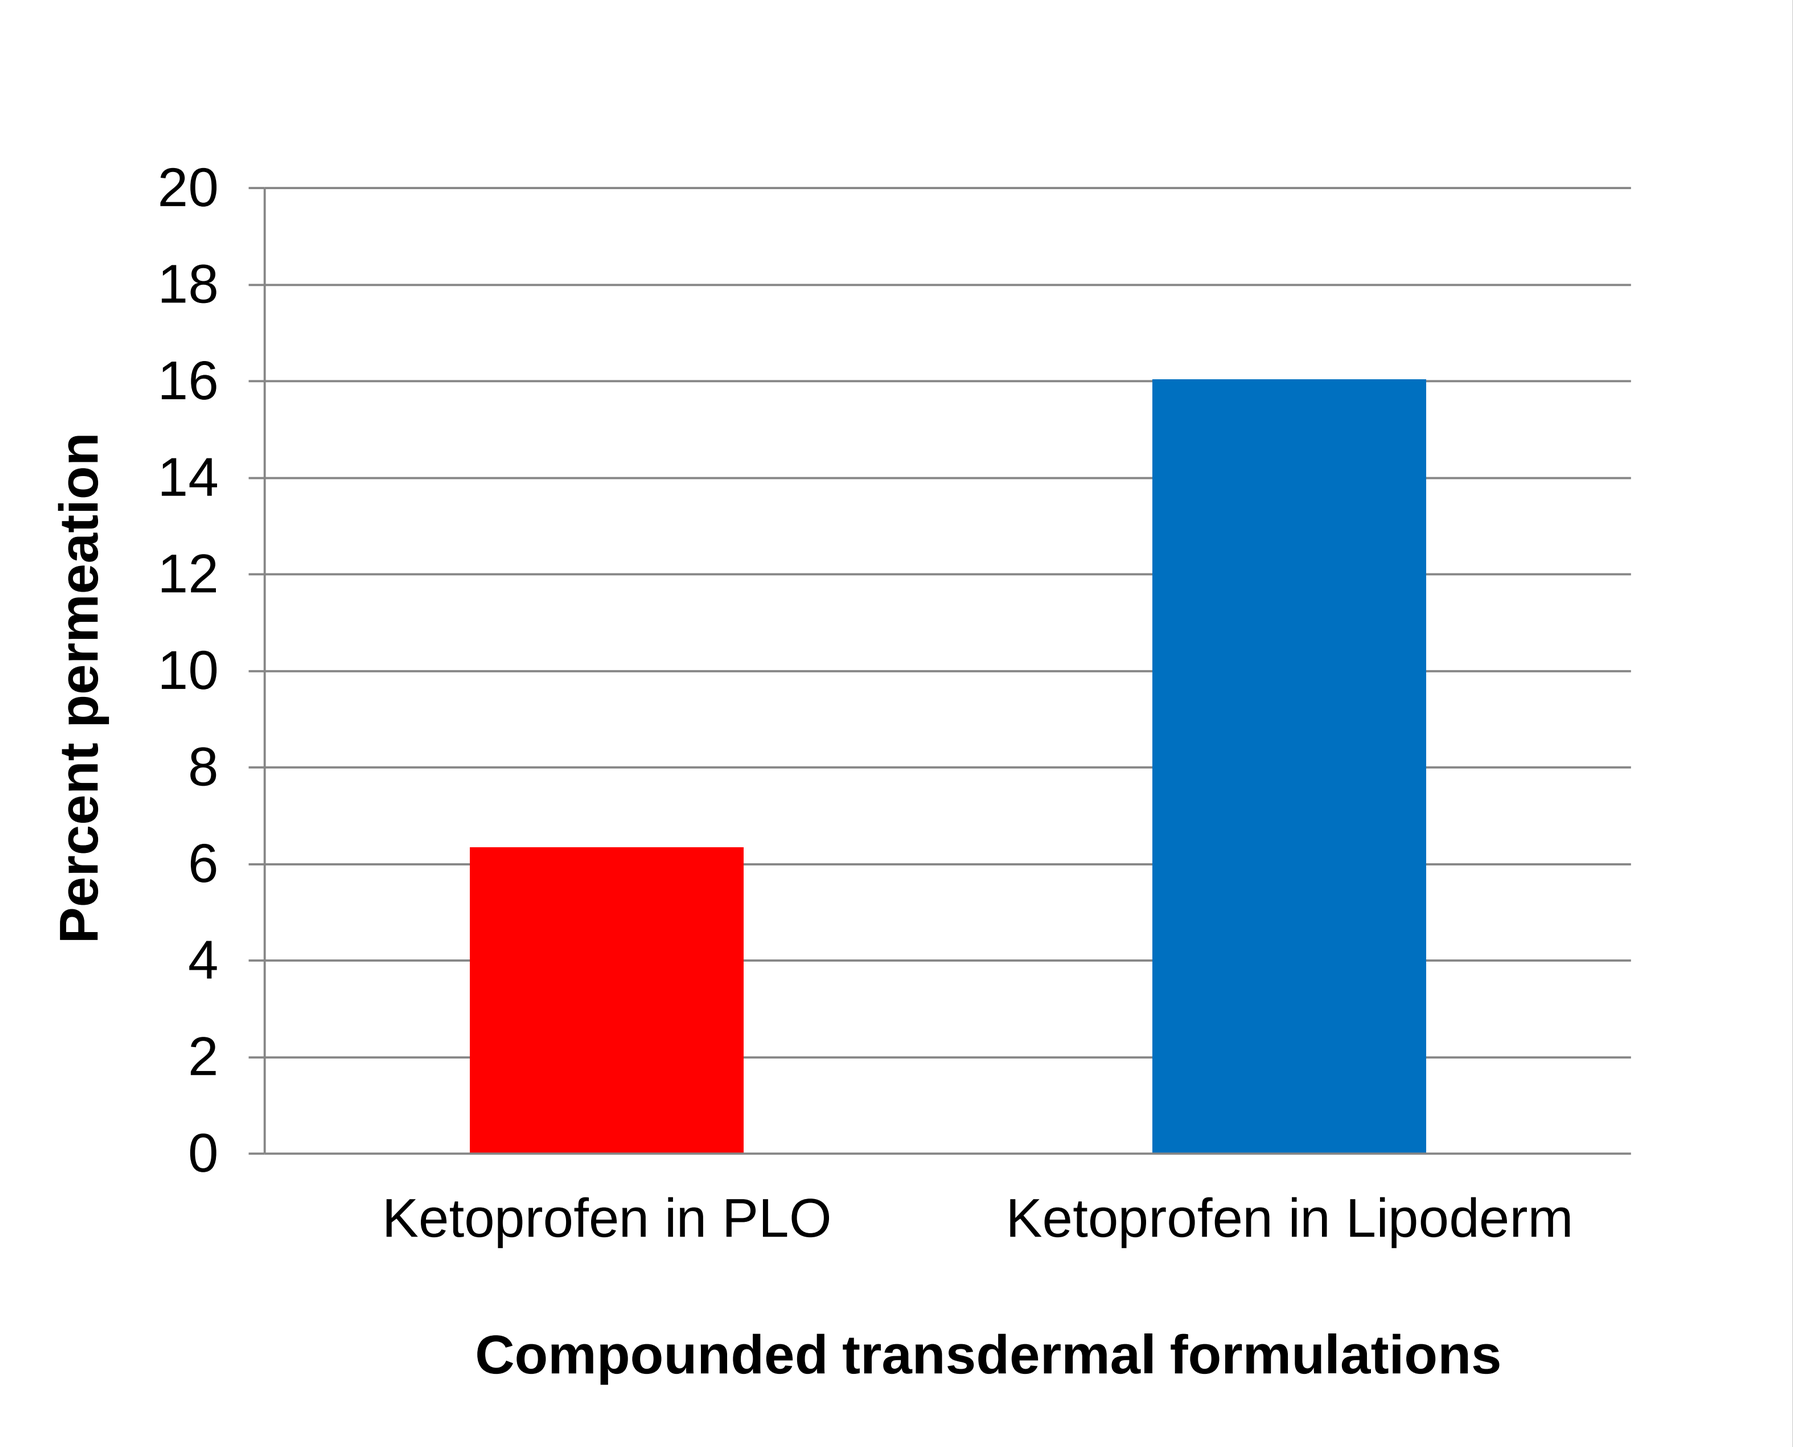
20
18
16
14
12
10
8
6
4
2
0
Percent permeation
Ketoprofen in PLO
Ketoprofen in Lipoderm
Compounded transdermal formulations

Figure 3.
Percent permeation of ketoprofen through the stratum corneum

1. Franz TJ, Lehman PA, Raney SG: Use of excised human skin to assess the bioequivalence of topical products. *Skin Pharmacol Physiol* 22: 276–286, 2009.
2. Schnitzer TJ: Update on guidelines for the treatment of chronic musculoskeletal pain. *Clin Rheumatol* 25 (Suppl 1): 22-29, 2006.
3. Williams KD: Managing pain with transdermal ketoprofen. *Int J Pharm Compd* 14: 204-206, 2010.

*Please send correspondence to: PCCAScience@pccarx.com

For a copy of this poster, please scan the QR code